Information transfer rate exceeds 200 bits/second in a high-density intracortical brain-computer interface

Paradromics

Sean M. Perkins†, Michael Trumpis†, Michael E. Reitman, Beata Jarosiewicz, Aashish N. Patel, Adam Weiss, Jacob W. Scott, Kurtis Nishimura, Matthew R. Angle, Shaoyu Qiao‡, and Vikash Gilja‡

LBP040.03

Standard for Optimizing Neural Interface Capacity (SONIC)

BRAIN-COMPUTER INTERFACE (BCI) DESIGN REQUIREMENTS: SAFETY | DURABILITY | **PERFORMANCE**

THE NEED FOR BCI HARDWARE BENCHMARKS

- **Rapid iteration:** Benchmarks provide objective metrics that guide device design. Iteration can occur more rapidly in a preclinical environment.
- **Informed decisions:** Standardized comparisons of BCI technologies enable patients, clinicians, and researchers to choose the BCI that best meets their needs.

FUNDAMENTAL BCI SPECIFICATIONS

- **Information Transfer Rate (ITR):** Measurement (in bits/second) of the speed and accuracy of decoding.
- **Intrinsic delay ($t_{intrinsic}$):** Delay between neural activity and decoded action due to measurement, signal processing, and algorithmic choices.

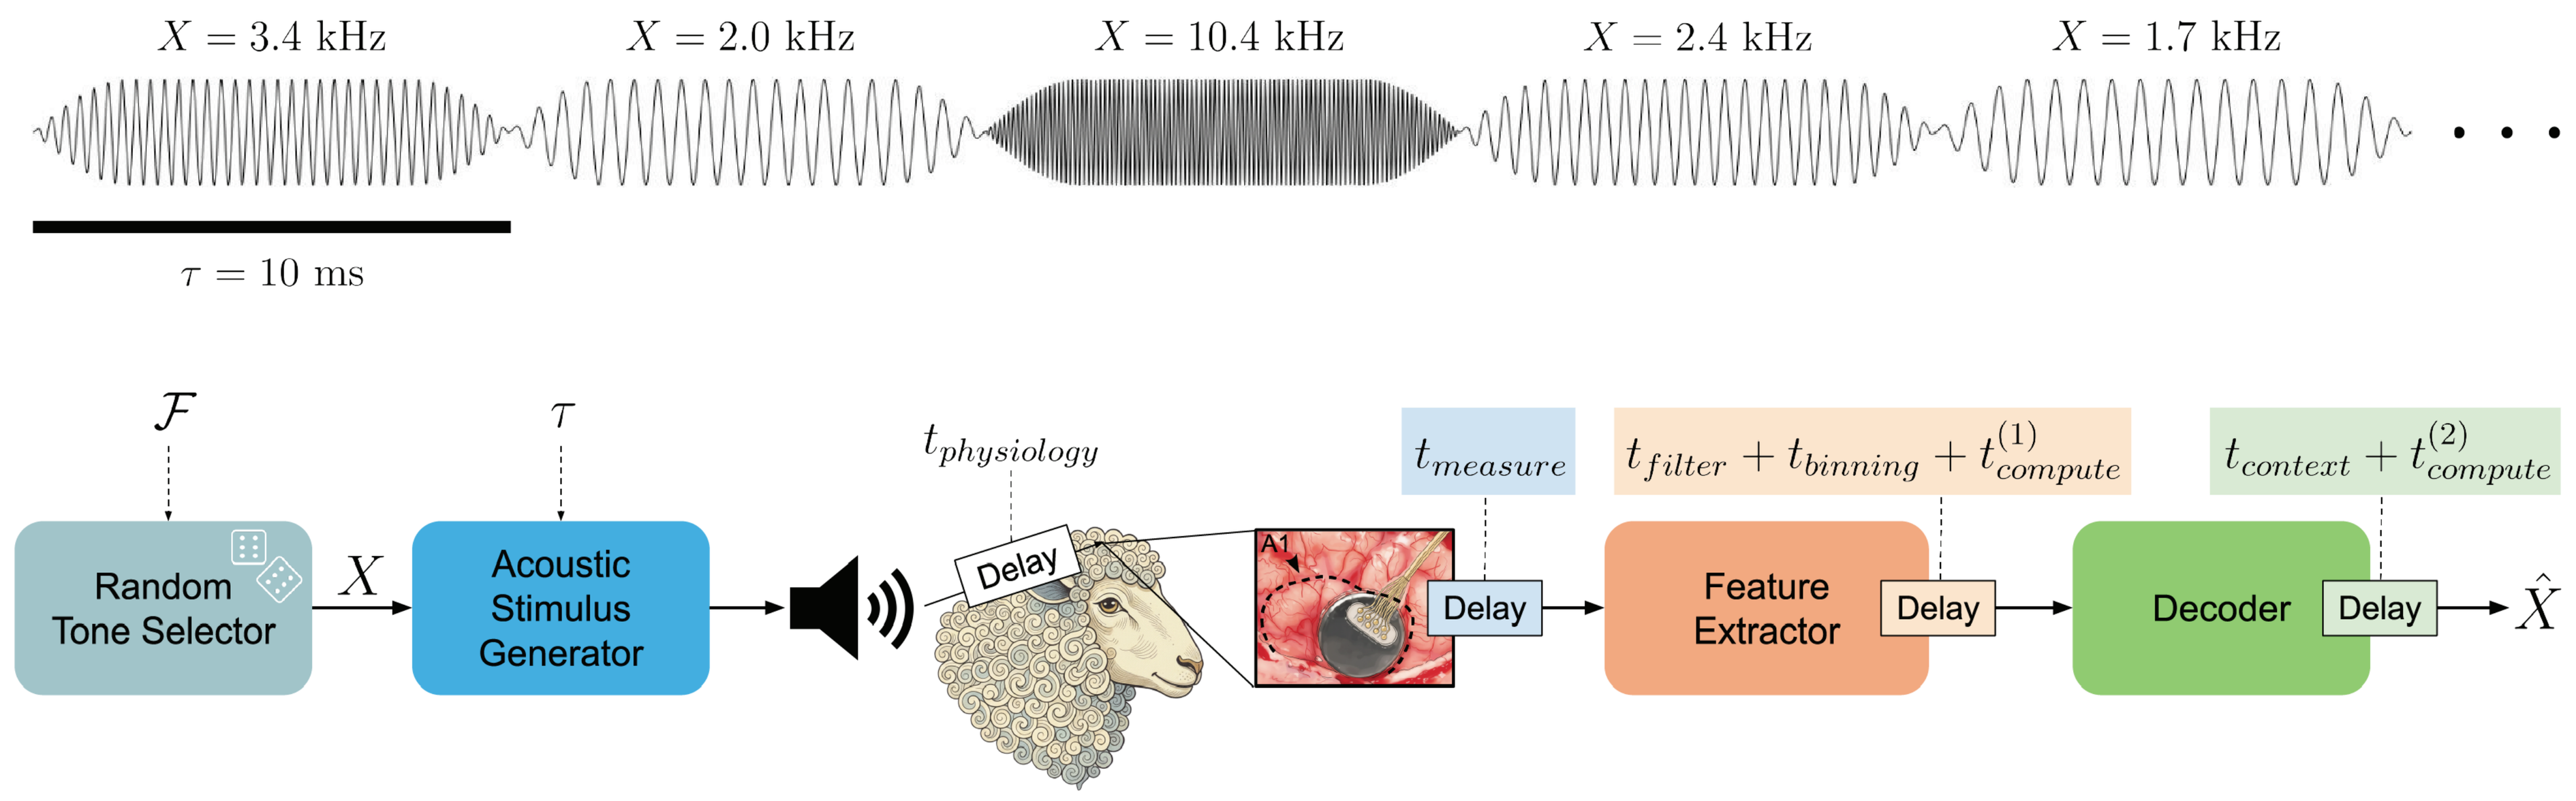

X = 3.4 kHz
X = 2.0 kHz
X = 10.4 kHz
X = 2.4 kHz
X = 1.7 kHz
τ = 10 ms
Random Tone Selector
Acoustic Stimulus Generator
Delay
Feature Extractor
Decoder

$$ITR = \frac{I_{corrected}(X;\hat{X})}{\tau} \qquad t_{intrinsic} = t_{measure} + t_{filter} + t_{binning} + t_{context} \qquad t_{bci} = t_{intrinsic} + t^{(1)}_{compute} + t^{(2)}_{compute}$$

SONIC is designed to avoid common ITR bottlenecks

POTENTIAL FACTORS	DESIGN CONSIDERATIONS
Physical neural interface, readout, and control electronics	• **Auditory cortex is accessible by many devices** • **Tone duration (τ) matched to temporal resolution of device**
Signal processing and decoding	• Unconstrained to allow algorithms to be tailored to the properties of each device
Complexity and timing of stimuli	• Neurons in auditory cortex encode auditory stimuli • Auditory stimuli can have a high ITR (i.e. not the limiting factor)
Responsiveness of neurons at the implant site to the information	• Neural dynamics in auditory cortex are fast • Frequencies ($\mathcal{F}$) matched to tonotopy at implant site

Exploratory Sessions with Connexus BCI

Dense frequencies good for sheep C | Refined frequencies needed for sheep E | ITR peaks around 10 ms tone duration

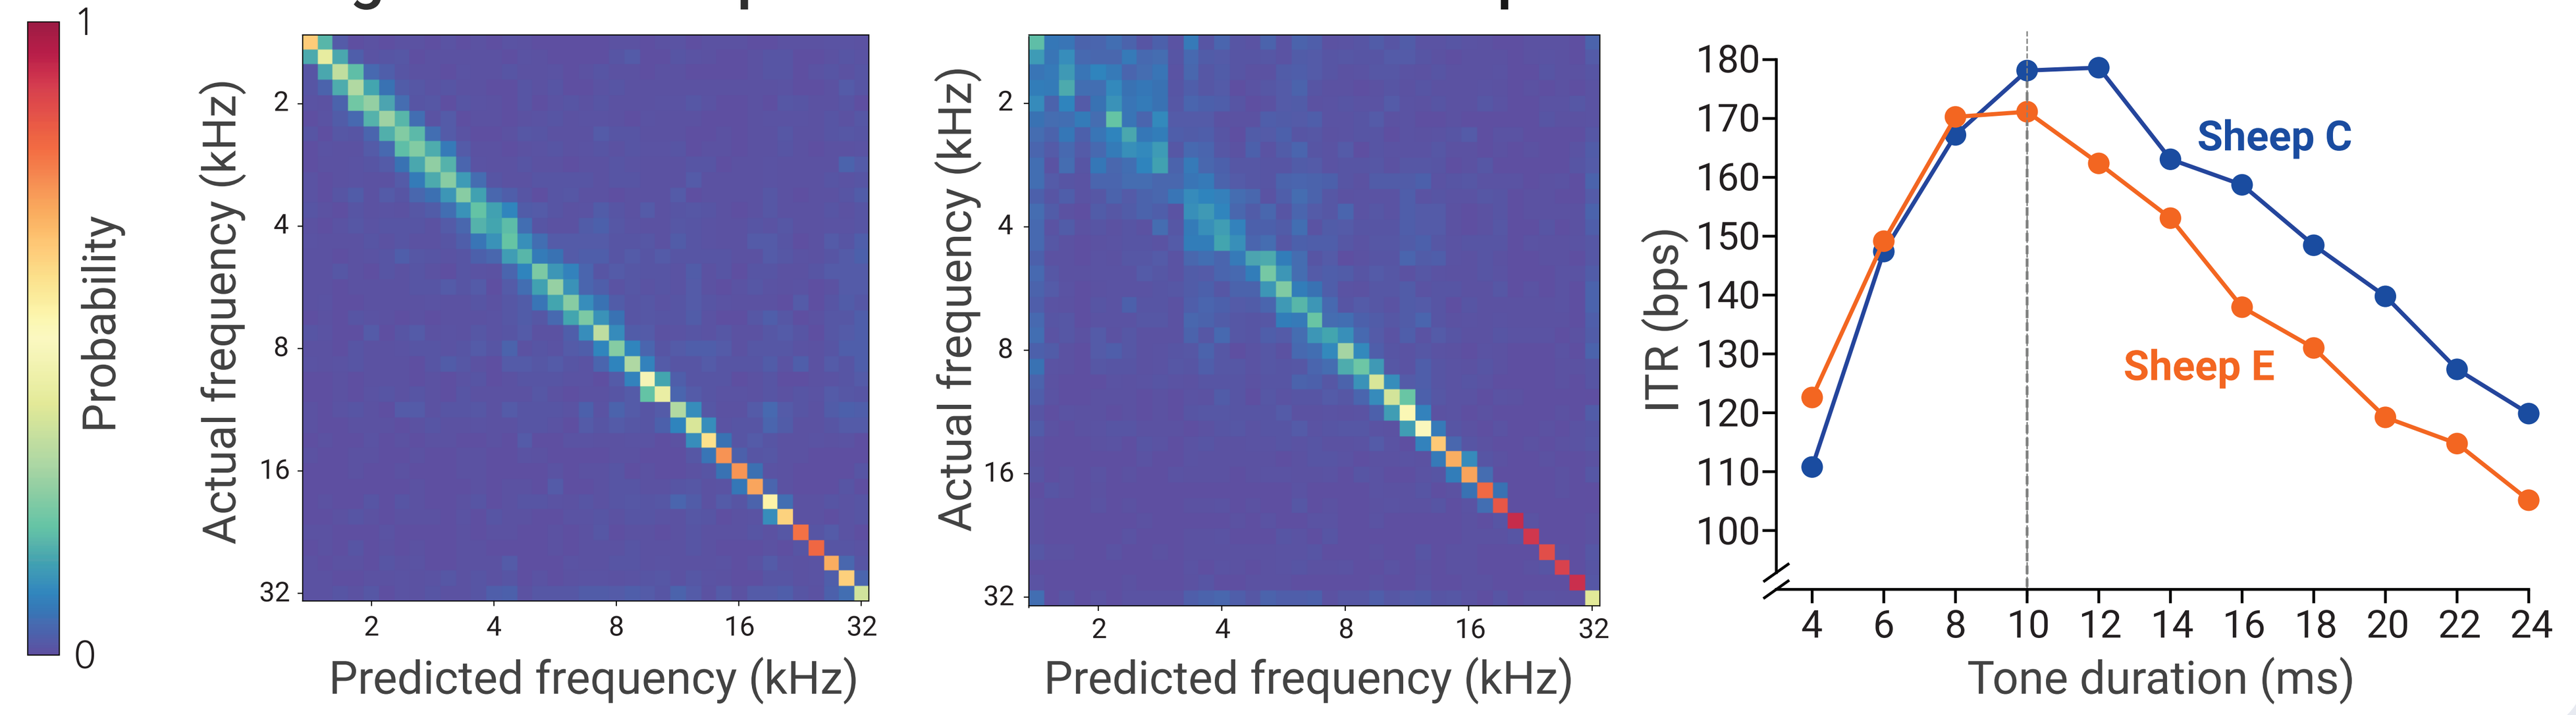

Probability
Actual frequency (kHz)
Predicted frequency (kHz)
ITR (bps)
Sheep C
Sheep E
Tone duration (ms)

Benchmarking Sessions with Connexus BCI

ITR exceeds 200 BPS with intrinsic delay of 56 ms

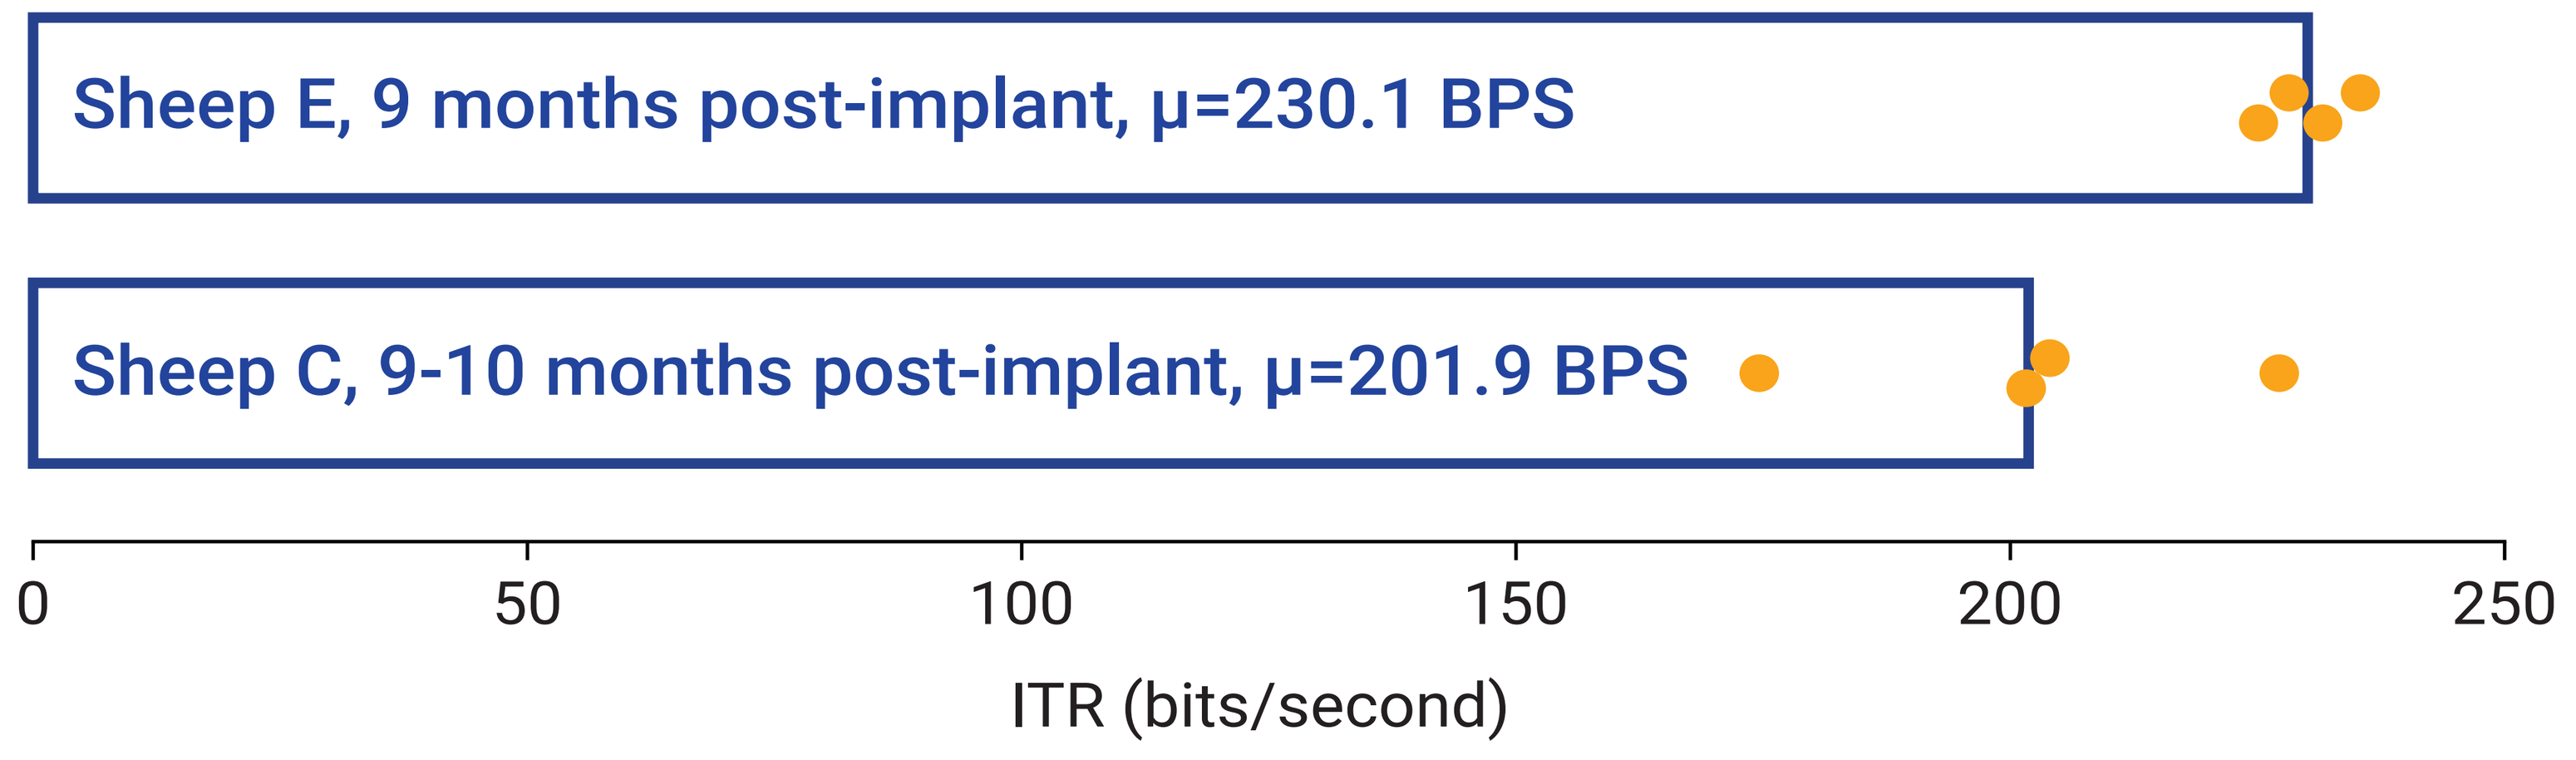

Sheep E, 9 months post-implant, μ=230.1 BPS
Sheep C, 9-10 months post-implant, μ=201.9 BPS
ITR (bits/second)

- **Features:** spike counts & spike-band power (421 chan) in 5 ms bins 10-60 ms after tone onset
- **Decoder:** two-layer 1D CNN (convolutional neural network)
- **Evaluation:** k-fold cross-validation ensuring no feature overlap across folds

- **Intrinsic delay: 56 ms**
 - Measurement delay: ~1 ms
 - Filter delay: 5 ms
 - Binning delay: 5 ms
 - Context delay: 45 ms

ITR exceeds 100 BPS with intrinsic delay of 6 ms

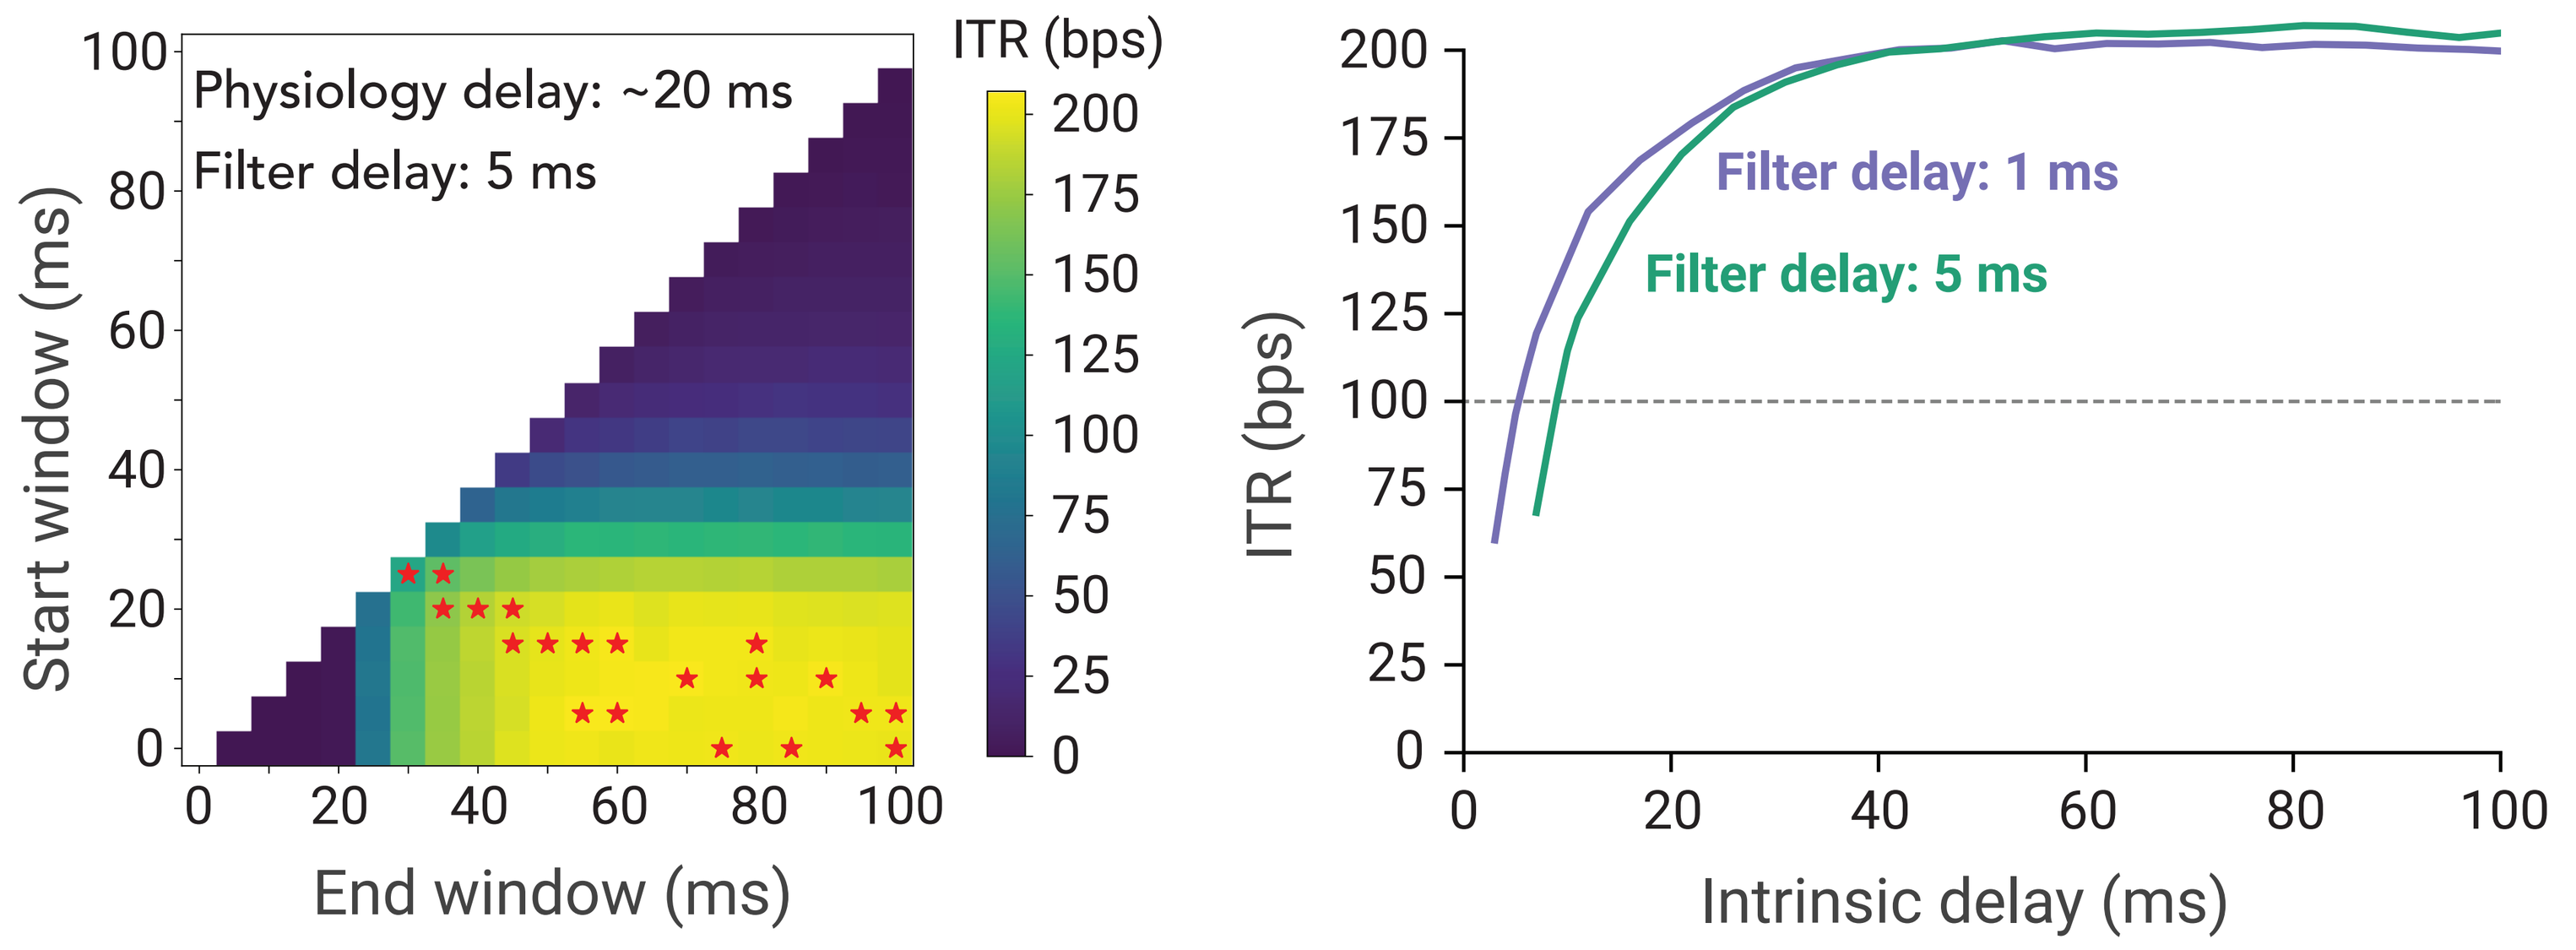

Physiology delay: ~20 ms
Filter delay: 5 ms
Start window (ms)
End window (ms)
ITR (bps)
Filter delay: 1 ms
Filter delay: 5 ms
Intrinsic delay (ms)

High ITR Depends on Channel Count

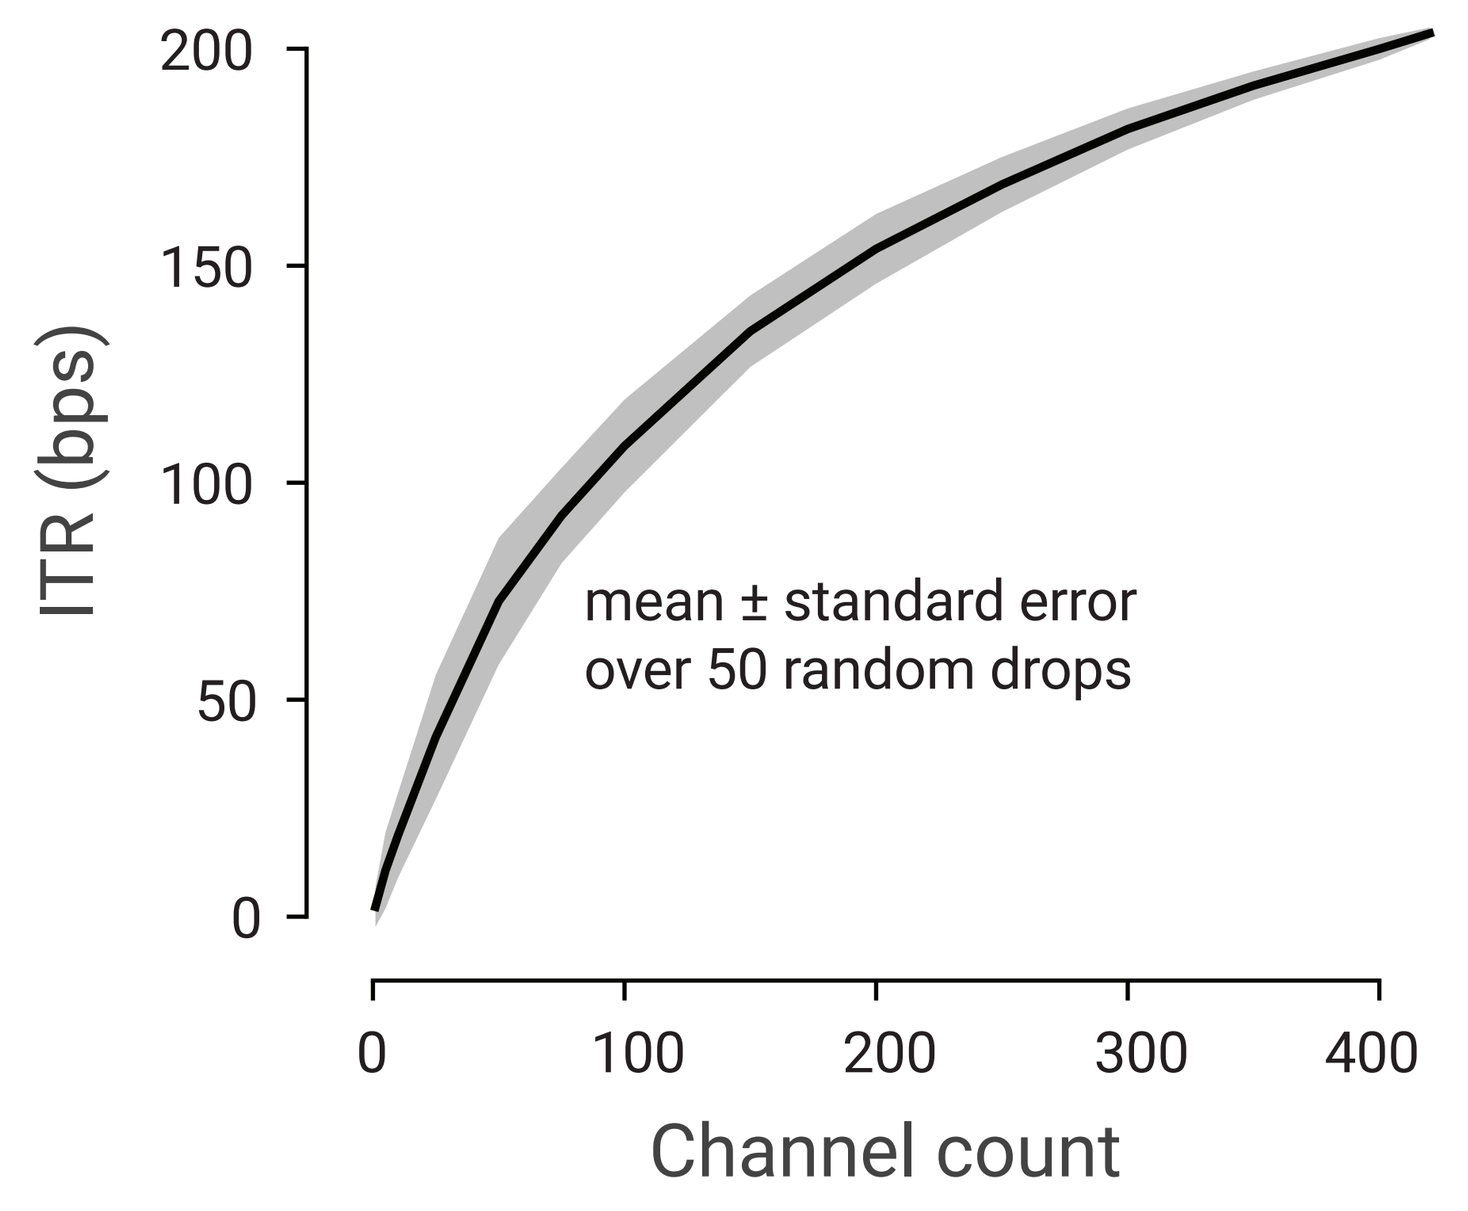

ITR (bps)
mean ± standard error over 50 random drops
Channel count

INTERACTIONS

- Implant anatomy
- Number of arrays
- Electrode pitch
- Cortical coverage
- Spatial homogeneity of neural activity

Sheep C example (204 BPS) | Sheep E example (232 BPS)

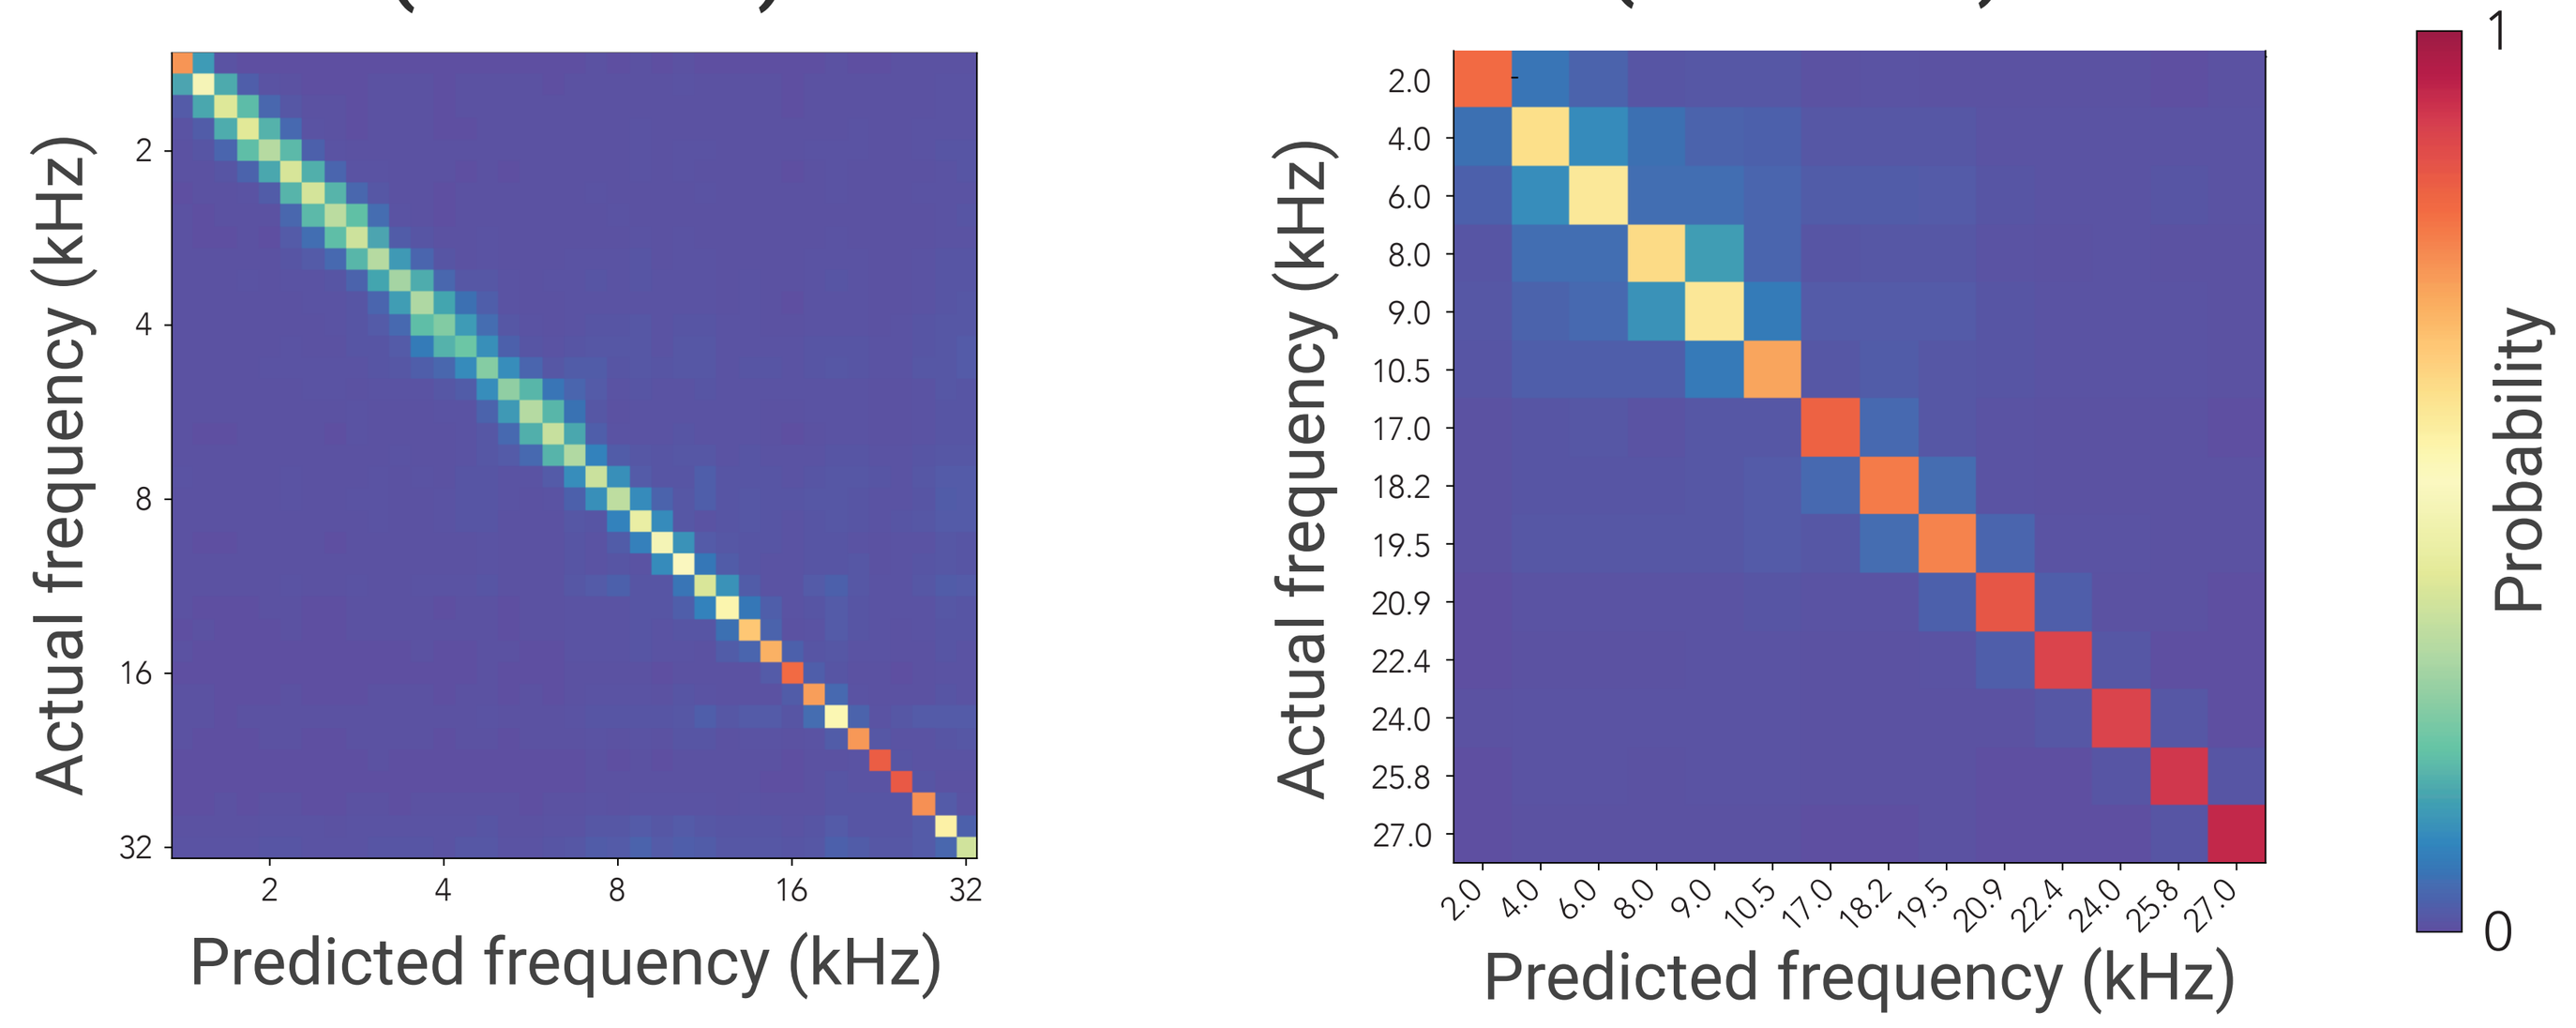

Actual frequency (kHz)
Predicted frequency (kHz)
Probability

ITR and Delay Determine Viability of Applications

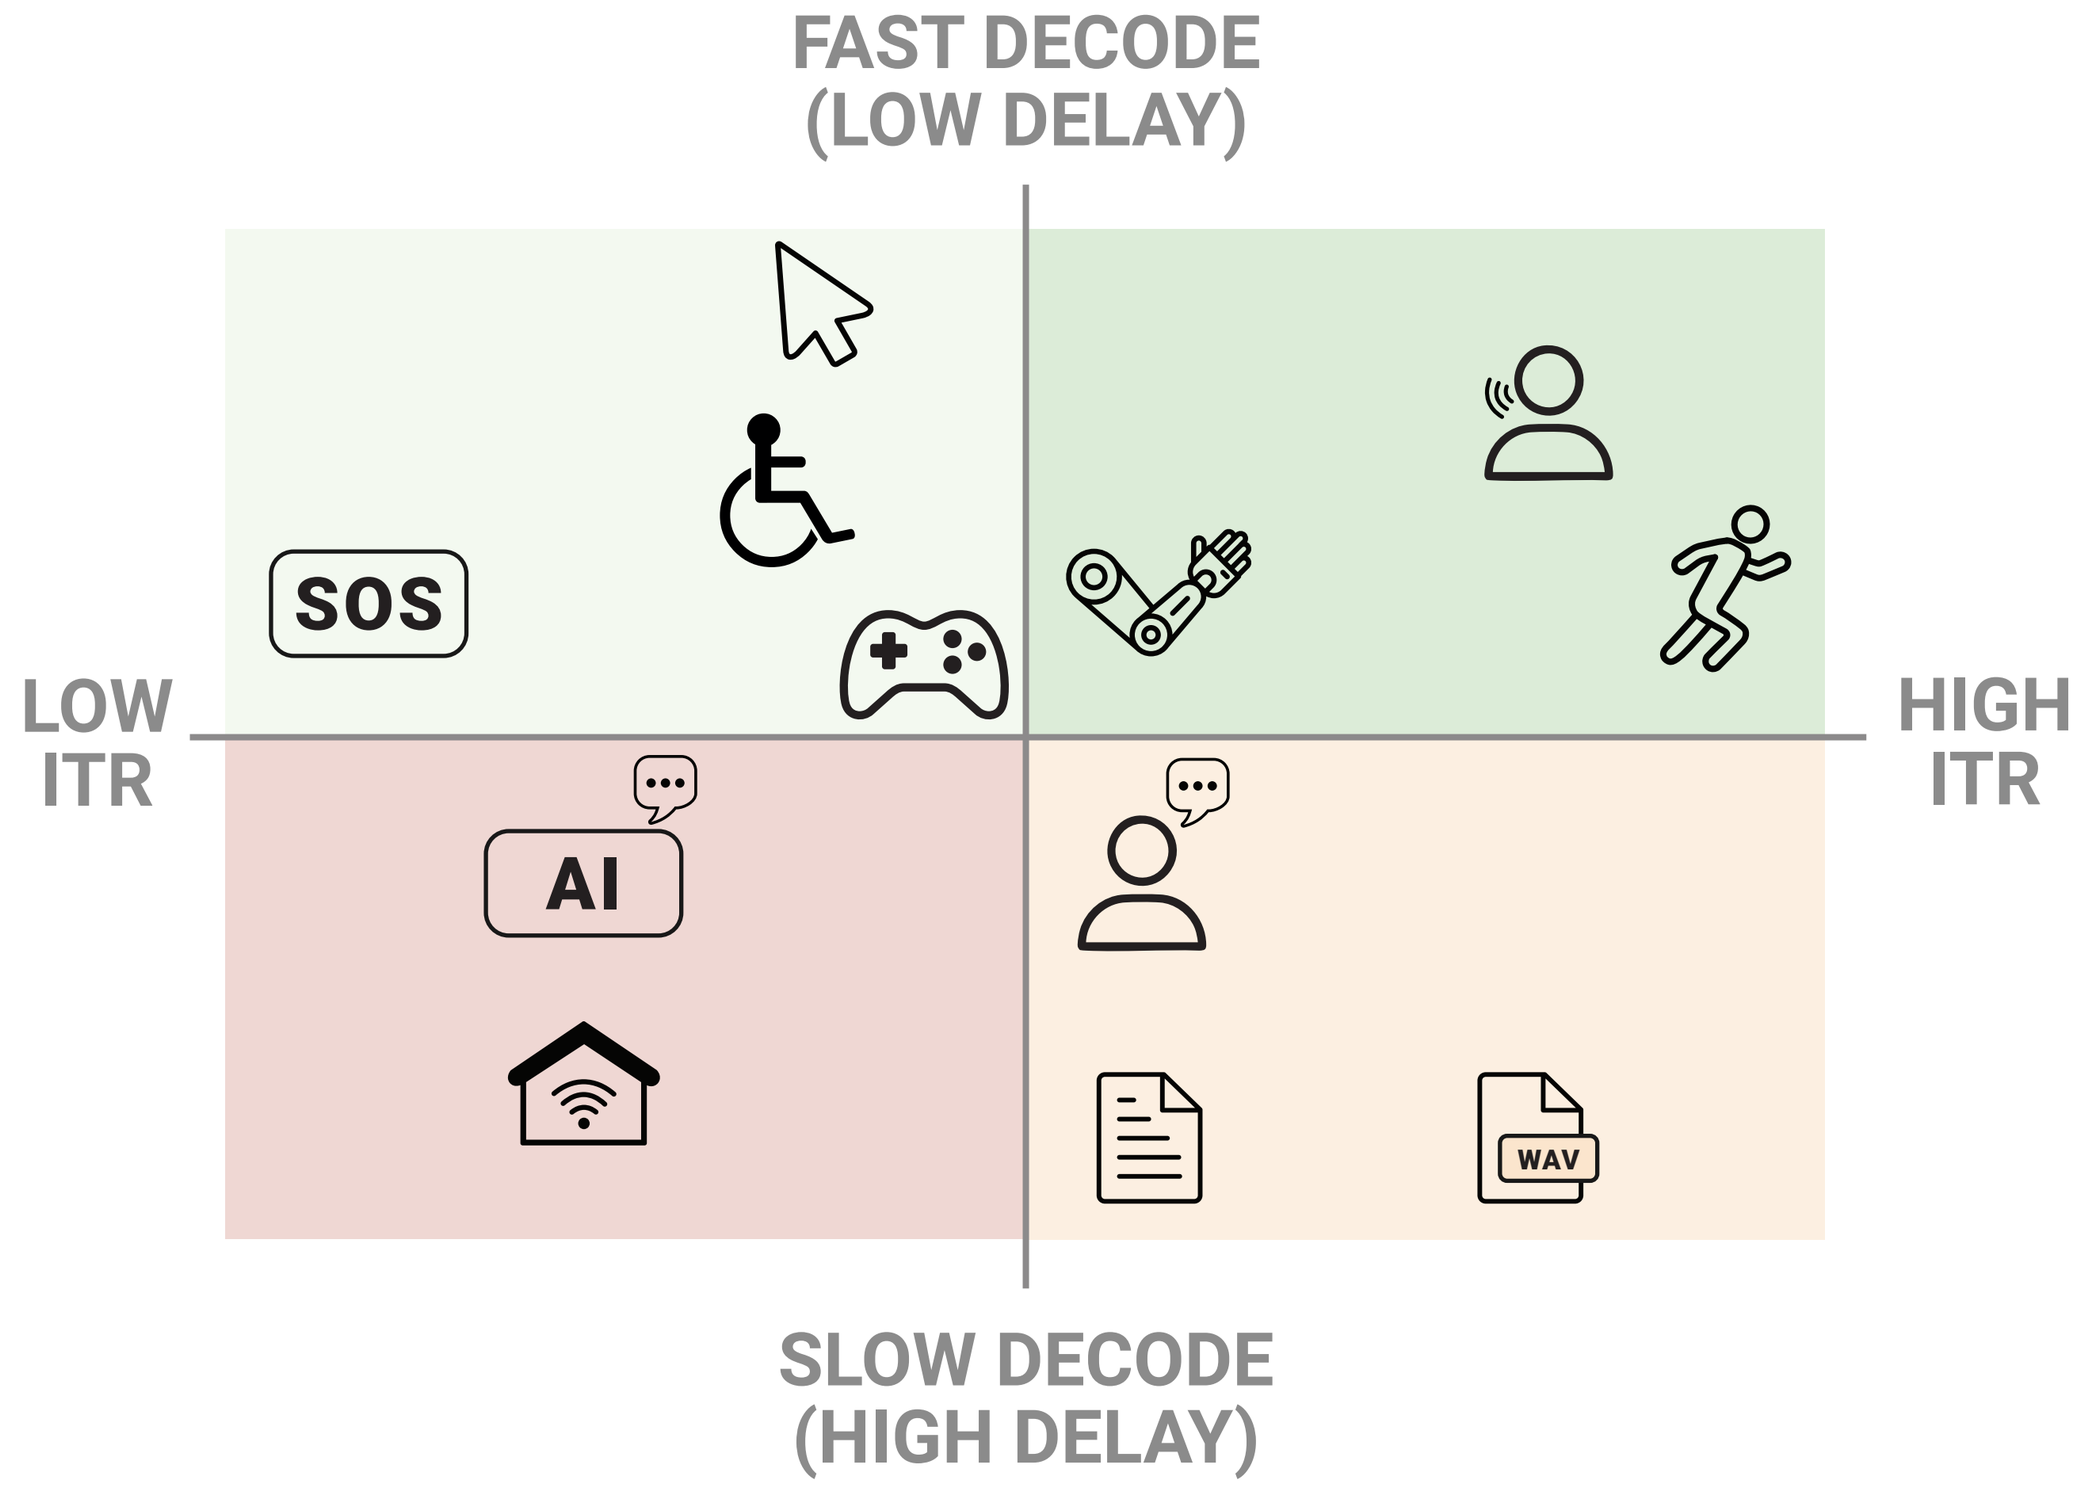

FAST DECODE (LOW DELAY)
LOW ITR
HIGH ITR
SLOW DECODE (HIGH DELAY)
SOS
AI
WAV

ITR CAPACITY BEYOND APPLICATION MINIMUMS ENABLES:

- Delay reduction
- Error reduction
- Sensitivity to application-relevant signals
- Multitasking
- Auxiliary information decoding (e.g. context, error signals, mood, affect)

Early Feasibility IDE Study

RESTORING COMMUNICATION WITH THE CONNEXUS BCI SYSTEM: **SAFETY** | DURABILITY | PERFORMANCE

INTENDED PARTICIPANT POPULATION

- Progressive neuromuscular disease (i.e. ALS) or neural injury (i.e. stroke)
- Severe dysarthria or anarthria

ENDPOINTS

- **Primary:** Safety
- **Secondary:** Clinical Outcome Assessments, Patient Reported Outcomes, speech BCI performance

STUDY SITES & INVESTIGATORS

- UC Davis (Study PI: David Brandman, M.D., Ph.D.)
- Massachusetts General Hospital (Daniel Rubin, M.D., Ph.D.)
- University of Michigan Health (Matthew Willsey, M.D., Ph.D.)

INITIAL APPLICATIONS

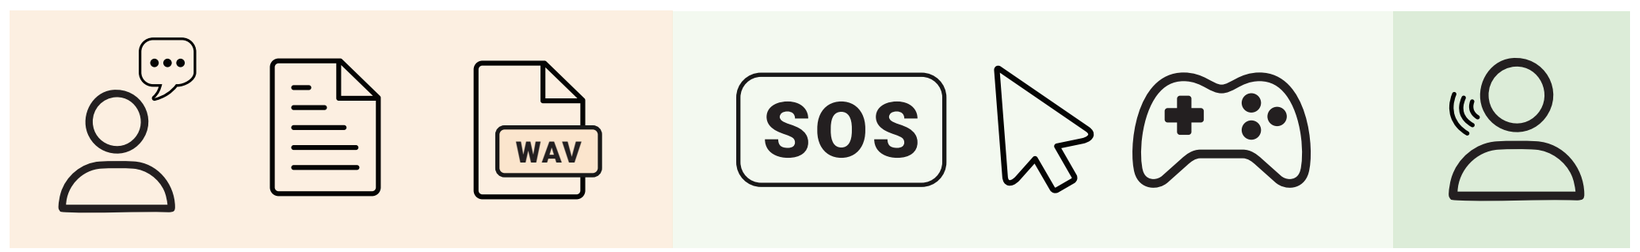

WAV
SOS

Anticipated study launch: End of 2025 (pending regulatory approval)

Conclusion

Using the SONIC preclinical benchmark, the Connexus BCI system has achieved an ITR > 200 bps, the highest reported BCI ITR to date. This far exceeds the ITR estimated for other types of neural interfaces (e.g. ECoG: ~10 bps; endovascular ~1 bps).

FUTURE DIRECTIONS

- Signal processing & algorithmic optimizations
- Electrode density and cortical coverage
- Channel count and sampling rate
- Control for attentional fluctuations
- Complex stimuli (e.g chords, naturalistic sounds)

PREPRINT

science@paradromics.com

Acknowledgments

Current and former Paradromics employees for their contributions to the design, manufacturing, and testing of Connexus BCI. Our interns—Surya Pandiaraju, Tereza Okálová, Nathan Liu, and Yusuf Ali—for their helpful technical discussions. Veterinary staff at the animal facility for their support with animal preparation and care.

†Equal contribution ‡*Equal contribution as senior authors*